Effect of Once-Daily Bromfenac Ophthalmic Solution on Photophobia Associated with Cataract Surgery

DE Beahm, MD[1], RL Berry, MD[2], L Cacioppo, MD[3], R Bianca, PhD[4], JA Gow, MD[4], TR McNamara, PharmD[4] for the Bromfenac Ophthalmic Solution Once Daily Study Group

[1]Beahm Eye Center, Great Bend, KS, [2]Eye Care Arkansas, PA, Little Rock, AR, [3]Hernando Eye Institute, Brooksville, FL, [4]ISTA Pharmaceuticals®, Inc., Irvine, CA

ABSTRACT

Purpose: To evaluate the effect of bromfenac ophthalmic solution administered once daily on photophobia associated with cataract surgery.
Methods: Subjects were assigned randomly to receive either bromfenac ophthalmic solution QD (n=360) or placebo QD (n=182). Dosing began 1 day before cataract surgery and continued through post-surgery Day 14. Photophobia was rated on a four point (0 to 3) scale. Rating was to be completed by the subject within 1 hour after instillation of test agent into the study (operative) eye and recorded in a diary.
Results: The proportion of subjects who were free of photophobia was significantly greater in the bromfenac treatment group than in the placebo treatment group by Day 1 (62% vs 53%, p<0.05), Day 3 (78% vs 65%, p<0.01), Day 8 (86% vs 69%, p<0.0001), and Day 15 (90% vs 73%, p<0.0001). The mean photophobia score was significantly less in the bromfenac treatment group than in the placebo treatment group at Day 1 (0.5 vs 0.7, p<0.01), Day 3 (0.4 vs 0.8, p<0.0001), Day 8 (0.2 vs 0.8, p<0.0001), and Day 15 (0.2 vs 0.7, p<0.0001).
Conclusion: Bromfenac ophthalmic solution QD was effective for the treatment of photophobia associated with cataract surgery.

INTRODUCTION

- Bromfenac is a non-steroidal anti inflammatory drug (NSAID) that works by blocking the production of prostaglandins, mediators of systemic and localized (e.g., ocular) inflammation.[1]
- Bronuck® (bromfenac sodium ophthalmic solution 0.1%) was approved in Japan in May 2000 for the treatment of patients with blepharitis, conjunctivitis, scleritis (including episcleritis) and post-operative inflammation.[2-9]
- Xibrom™ (bromfenac ophthalmic solution 0.09%), administered twice daily, was approved by the Food and Drug Administration (FDA) in March 2005 for the treatment of patients with post-operative ocular inflammation and in January 2006 for the treatment of patients with post-operative ocular pain.10
- Through December 2005, with more than 10 million uses of Bronuck in Japan and more than 204,000 uses of Xibrom in the U.S.,:[11-12]
 - no serious systemic drug-related adverse event was reported, and
 - the incidence of spontaneously reported serious ocular adverse events was <0.0000025%.
- Based on extensive post-marketing experience and data from clinical trials, bromfenac ophthalmic solution has demonstrated an excellent safety profile.
- Bromfenac ophthalmic solution QD is a new formulation of topical bromfenac to be administered once daily and is currently not approved by the FDA. This formulation should be more convenient for patients, should improve compliance with the treatment regimen, and should enhance clinical efficacy.

PURPOSE

- **To evaluate the effect of bromfenac ophthalmic solution administered once daily on photophobia associated with cataract surgery.**

SUBJECTS AND METHODS

- Multi-center, randomized, double-masked, parallel group, placebo (vehicle)-controlled study.
- Data integrated from two separate studies conducted under a common protocol.
- Subjects assigned randomly to receive either bromfenac QD (n=360) or placebo QD (n=182).
- Dosing began 1 day before cataract surgery and continued through post-surgery day 14.

ENROLLMENT CRITERIA

Key Inclusion Criteria

- Males or females ≥ 18 years of age.
- Unilateral cataract surgery (phacoemulsification or extracapsular) with posterior chamber intraocular lens implantation and no plans for other ophthalmic procedures (e.g., relaxing incisions, iridectomy, conjunctival excisions, etc) during surgery.
- Best Corrected Visual Acuity of at least 20/200 in the non-study eye.
- Negative urine pregnancy test for women capable of becoming pregnant.
- Informed consent approved by Institutional Review Board or Independent Ethics Committee.
- IOP ≥ 5 mmHg and ≤ 22 mmHg in study eye.

Key Exclusion Criteria

- Known hypersensitivity to bromfenac or to any component of the test agents and “procedural” medications such as anesthetic and/or fluorescein drops.
- Known hypersensitivity to salicylates or other NSAIDs.
- Intraocular inflammation (i.e, anterior chamber cells or flare on slit lamp examination) or ocular pain in either eye at screening.
- Uncontrolled active or chronic/recurrent ocular or systemic disease likely to affect wound healing (e.g., diabetes mellitus, systemic connective tissue disease, severe atopic disease).
- Blood dyscrasia, bone marrow suppression, uncontrolled/unstable peptic ulcer disease, inflammatory bowel disease, ulcerative colitis, or uncontrolled/unstable pulmonary, cardiac, vascular, autoimmune, hepatic, renal, or central nervous system disease.
- Use of anticoagulants ≤7 days before initiating treatment with test agent, need for anticoagulant therapy (including aspirin >165 mg/day), or known or suspected bleeding disorder.
- Use of ocular, topical, or systemic NSAIDs or gentamicin, or cyclosporine ophthalmic emulsion ≤7 days before initiating treatment with test agent.
- Use of ocular, topical, inhaled, or oral corticosteroids ≤15 days before initiating treatment with test agent, or depo-corticosteroids ≤45 days before initiating treatment with test agent.
- Use of tamsulosin (Flomax).
- Use of ocular, topical, or systemic medication (e.g., antihistamines, mast cell stabilizers, ocular prostaglandins, regular use of preserved artificial tears, etc.) that could interfere with lacrimation, wound healing, the test agent, or the interpretation of study results, ≤7 days before initiating treatment with test agent.
- Active corneal pathology or corneal scarring in either eye at screening (except mild superficial punctate keratitis in the non-study eye).
- Extraocular/intraocular inflammation in either eye at screening (blepharitis allowed if scurf only, and no concurrent conjunctivitis or lid erythema/edema) or ongoing, unresolved uveitis.
- Radial keratotomy, corneal transplant, or corneal refractive surgery in the study eye ≤2 years before initiating treatment with test agent.
- Participation in a study of an investigational drug or device ≤30 days before screening.

CRITERIA FOR EVALUATION:

1. Proportion of subjects who were free of photophobia.
 - Photophobia (light sensitivity) was rated on a four point (0 to 3) scale: 0 = none, 1 = mild, 2 = moderate, and 3 = severe.
 - Rating was to be completed by the subject within 1 hour after instillation of test agent into the study (operative) eye and recorded in a diary.
2. Safety: included adverse events and ophthalmologic exams

RESULTS

The proportion of subjects who were free of photophobia was significantly greater in the bromfenac treatment group than in the placebo treatment group by Day 1 (62% vs 53%, p<0.05), Day 3 (78% vs 65%, p<0.01), Day 8 (86% vs 69%, p<0.0001), and Day 15 (90% vs 73%, p<0.0001).

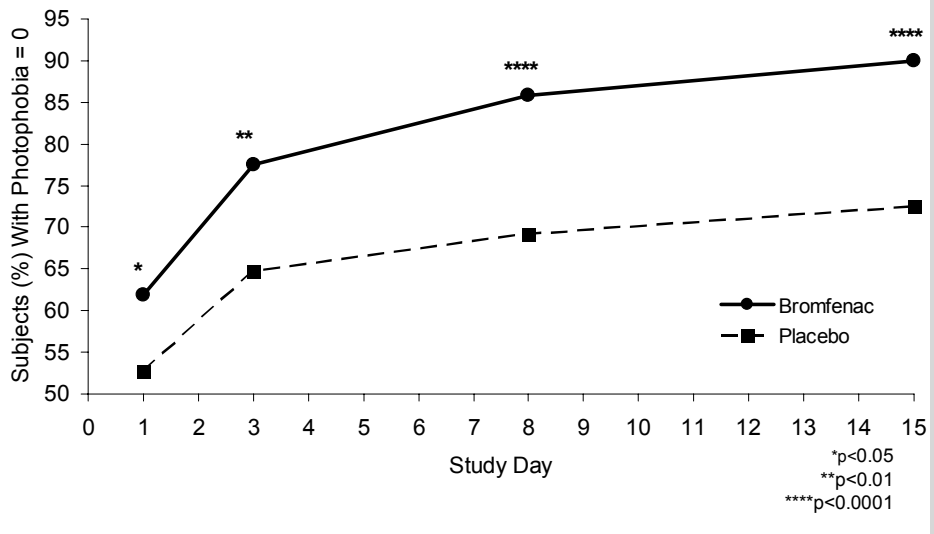
Subjects (%) With Photophobia = 0
95
90
85
80
75
70
65
60
55
50
*
**

Bromfenac
Placebo
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
Study Day
*p<0.05
**p<0.01
****p<0.0001

Figure 1. Photophobia Score: Subjects (%) with Grade 0 by Visit

The mean photophobia score was significantly less in the bromfenac treatment group than in the placebo treatment group at Day 1 (0.5 vs 0.7, p<0.01), Day 3 (0.4 vs 0.8, p<0.0001), Day 8 (0.2 vs 0.8, p<0.0001), and Day 15 (0.2 vs 0.7, p<0.0001).

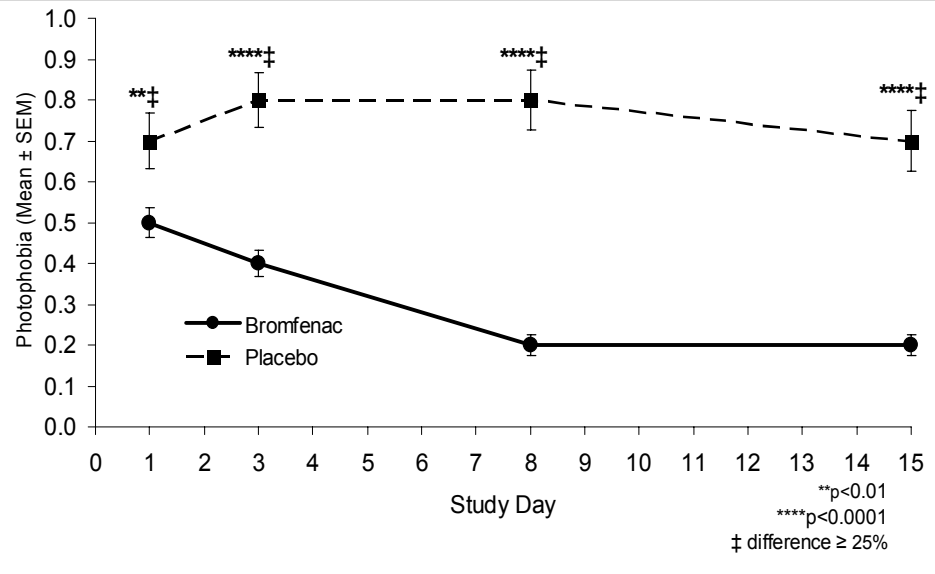
Photophobia (Mean ± SEM)
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
**‡
****‡
****‡
****‡
Bromfenac
Placebo
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
Study Day
**p<0.01
****p<0.0001
‡ difference ≥ 25%

Figure 2. Photophobia: Mean Grade at Each Visit

CONCLUSION

Bromfenac ophthalmic solution QD was effective for the treatment of photophobia associated with cataract surgery.

REFERENCES

1. Nichols J, Snyder RW. Topical nonsteroidal anti-inflammatory agents in ophthalmology. Curr Opin Ophthalmol 1998;9: 40-4.
2. Bronuck® package insert. Senju Pharmaceutical Co,. Ltd., Osaka, Japan, 2003.
3. Kawaguchi T, Kida T, Nemoto S, et al. Effect of bromfenac ophthalmic solution on ocular inflammation and corneal epithelial barrier function following cataract surgery. Folia Ophthalmol Jpn 2003;54: 76-9.
4. Ohara K, Ohkubo A, Miyamoto T, et al. Effect of bromfenac sodium on postoperative inflammation. Jpn J Cataract & Refractive Surgery 2004;18: 168-73.
5. Ohara K, Ohkubo A, Miyamoto T, et al. Prevention of miosis during cataract surgery by topical bromfenac sodium. Jpn J Clin Ophthalmol 2004;58: 1325-28.
6. Miyake-Kashima M, Takano Y, Tanaka M, et al. Comparison of 0.1% bromfenac sodium and 0.1% pemirolast potassium for the treatment of allergic conjunctivitis. Jpn J Ophthalmol 2004;48: 587-90.
7. Takamatsu F, Shioyama N, Soito Y, et al. Efficacy and adverse effects of bromfenac sodium following cataract surgery. Jpn H Clin Ophthalmol 2003;57:1233-37.
8. Kitao N, Shimoji H, Fukuda M. Post-marketing surveillance of bromfenac sodium (Bronuck®) ophthalmic solution. Jn. of the Eye 2005;22: 1299-1308.
9. Usui M, Masuda K. Effect of bromfenac sodium (AHR-10282B) eye drops on anterior uveitis. Ganka Rinsho Iho (J Rev Clin Ophthalmol) 1997;91: 39-44.
10. Xibrom™ package insert. ISTA Pharmaceuticals®, Inc., Irvine, California, 2006.
11. Donnenfeld ED, Donnenfeld A. Global experience with Xibrom (bromfenac ophthalmic solution) 0.09%: the first twice-daily ophthalmic nonsteroidal anti-inflammatory drug. Int Ophthalmol Clin. 2006;46:21-40.
12. Donnenfeld ED, Holland EJ, Stewart RH, et al. Bromfenac ophthalmic solution 0.09% (Xibrom) for postoperative ocular pain and inflammation. Ophthalmology. 2007;114:1653-62.

Financial support: ISTA Pharmaceuticals®, Inc., Irvine, CA, USA.

Financial disclosures: DEB, RLB, and LC were principal investigators. RB, JAG, and TRM are affiliated with ISTA Pharmaceuticals®, Inc.

Reference: Beahm DE, Berry RL, Cacioppo L, Bianca R, Gow JA, McNamara TR. Effect of once-daily bromfenac ophthalmic solution on photophobia associated with cataract surgery. Paper presented at: 34th Symposium on Cataract, IOL, and Refractive Surgery of the American Society of Cataract and Refractive Surgery; April 4-9, 2008; Chicago, IL. Abstract 412790.